CYTOTOXICITY EVALUATION OF A NOVEL LATANOPROST 0.005% NANOEMULSION

Melina del Papa[1], Julia Tau[2], Alejandro Aguilar[2], Alejandro Berra[2], María Silvia Passerini[1]

1) POEN Laboratory, Buenos Aires, Argentina. 2) Ocular Research Laboratory, Department of Pathology, Faculty of Medicine, University of Buenos Aires, Buenos Aires, Argentina.

BACKGROUND:

Glaucoma is one of the leading causes of blindness worldwide. One of the major risk factors for development and progression of glaucoma is elevated intraocular pressure (IOP). Currently, IOP reduction is the primary goal in glaucoma and ocular hypertension treatment in order to prevent visual field damage. PGF2α (Prostaglandin F2α) analogs are the most effective drugs for reducing IOP.

Most PGF2α analog products contain benzalkonium chloride (BAK) as a solubilizing and antimicrobial agent. However, there is a considerable amount of evidence of its deleterious effect on the ocular surface, especially in long-term treatments. The reported effects, both in *in vitro* and *in vivo* models, include dose-dependent and time-dependent toxicity to the corneal epithelium, the conjunctival epithelium, the stroma, and tear film constituents. BAK may also reduce epithelial cell integrity, impair healing, induce cytokine secretion, cause elevated production of conjunctival inflammatory cells, and reduce goblet cell number. Furthermore, BAK may impair tear function and reduce tear film stability through its effects on the integrity of the meibomian layer and disruption of the lipid film continuous multilayer structure, thus decreasing tear film break-up time. At cellular level, BAK not only induces growth arrest but also increases epithelial cell apoptosis and cytotoxicity.

A new latanoprost 0.005% nanoemulsion (LNe) was developed to improve patient comfort and tolerability. Nanotechnology allows to address new pharmaceutical forms that enable to formulate BAK-free latanoprost eye drops and improve thermodynamic stability of the formulation that make storage at room temperature possible.

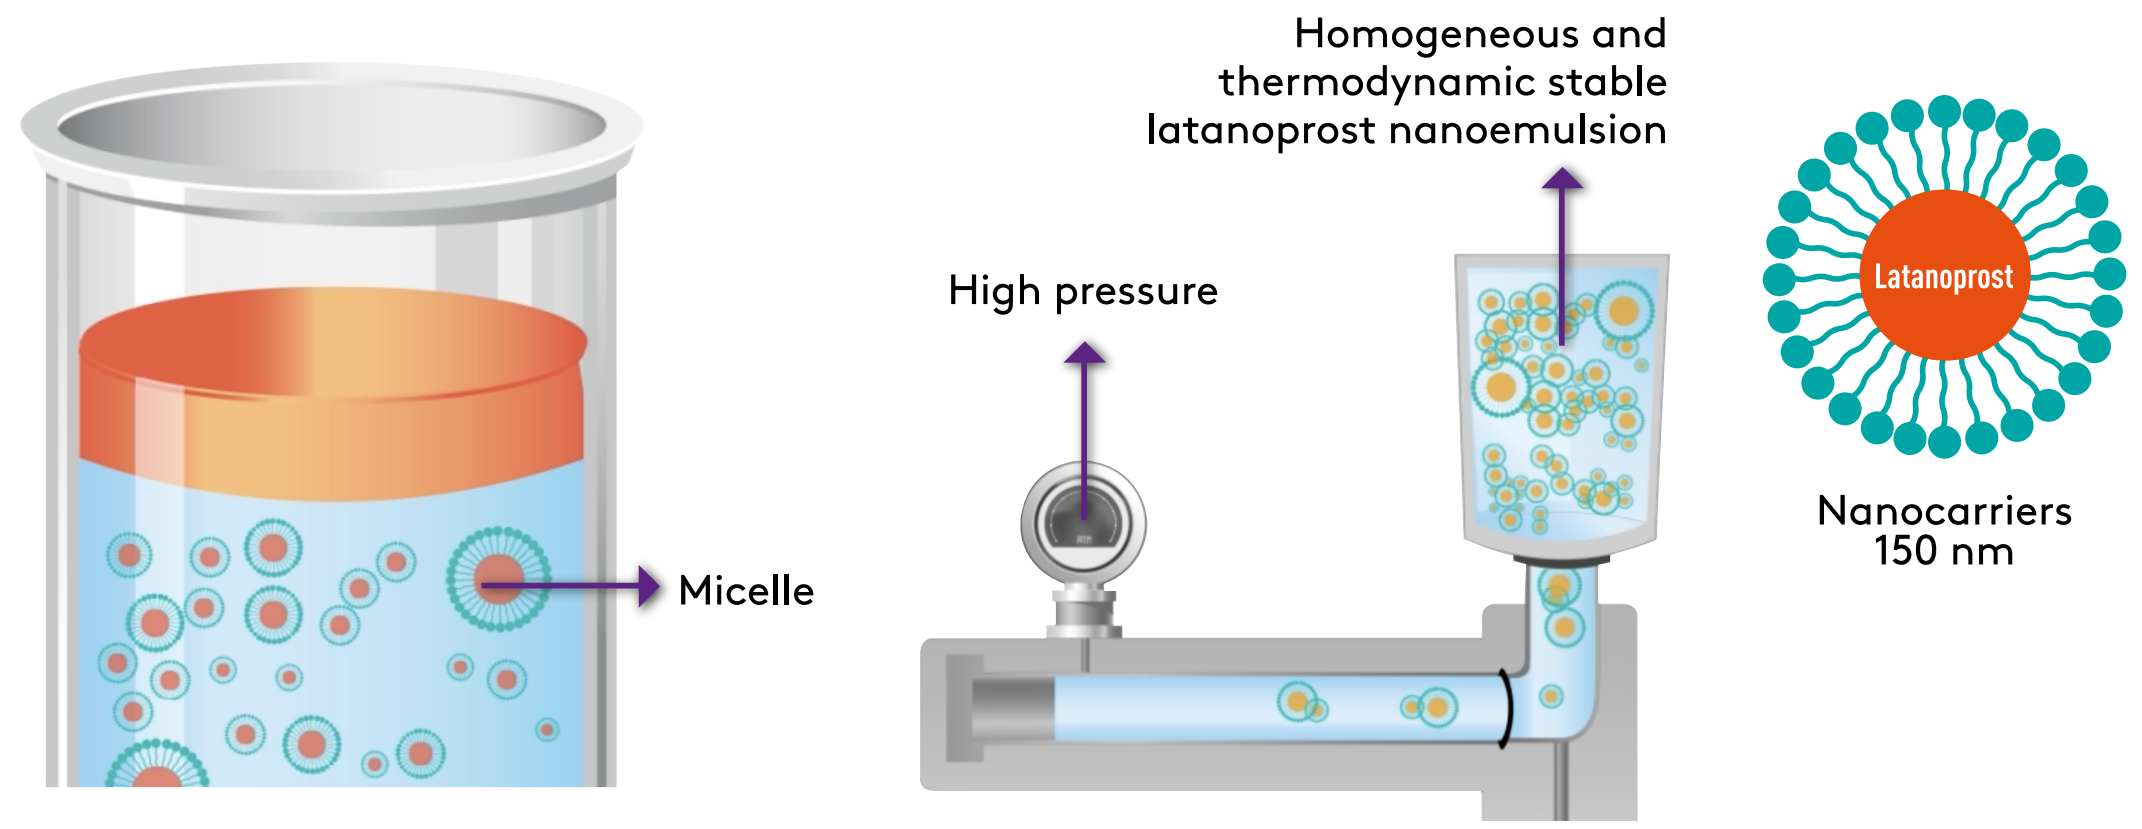
Homogeneous and thermodynamic stable latanoprost nanoemulsion
High pressure
Micelle
Latanoprost
Nanocarriers 150 nm

Figure 1. Obtention of latanoprost nanoemulsion by High Technology Microfluidizer.

PURPOSE:

To evaluate the cytotoxicity of a new Latanoprost BAK-free nanoemulsion, and to compare it with a Latanoprost solution containing BAK.

METHODS:

Human Conjunctival epithelial cells (IOBA-NHC) were incubated for 15, 30 and 60 minutes with 1:6 dilutions of either latanoprost solution containing BAK (LSc), (latanoprost 0.005%, BAK 0.02%; Xalatan, Pfizer) or latanoprost nanoemulsion (LNe), (latanoprost 0.005%, potassium sorbate 0.18%; Louten Emulsion, POEN).

Cell viability and proliferation:
Metabolic activity was measured as an indirect indicator of cell viability and proliferation by MTT assay. MTT is a yellow aqueous solution, which is reduced by dehydrogenases and other reducing agents present in metabolically active cells, to insoluble violet-blue formazan crystals. After dissolving formazan crystals, and incubation for 30 minutes, the absorbances of supernatants were measured at 570 nm.

Cell death evaluation:
Cell death was determined by flow cytometry using annexin V-FITC (AV) and propidium iodide-(PI) staining kit. AV binds to negatively charged phospholipid surfaces with a higher specificity for phosphatidylserine (PS). PS is an internal plasma membrane phospholipid that in the early apoptotic cascade is exposed on the outer layer of the plasma membrane. PI binds to nuclear components and can only enter the cell when it loses the integrity of its plasma membrane. The test described, discriminates viable cells (AV-/ PI-), early apoptotic cells (AV+/PI-) and late apoptotic on necrotic cells (AV+/PI+) *(See figure 2)*.

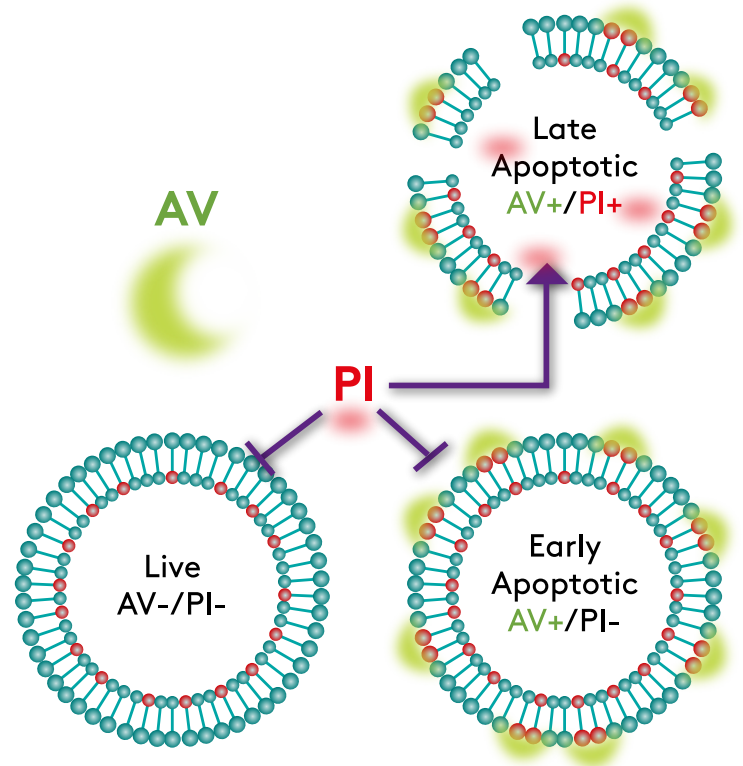
AV
PI
Late Apoptotic AV+/PI+
Live AV-/PI-
Early Apoptotic AV+/PI-

Figure 2. Annexin V-FITC and propidium iodide staining scheme.

RESULTS:

CELL VIABILITY AND PROLIFERATION

Values of cell viability and proliferation obtained from cells exposed to LNe were between 80 and 90% relative to control group, whereas values obtained from cells exposed to LSc were around 30% relative to control group at all study times.

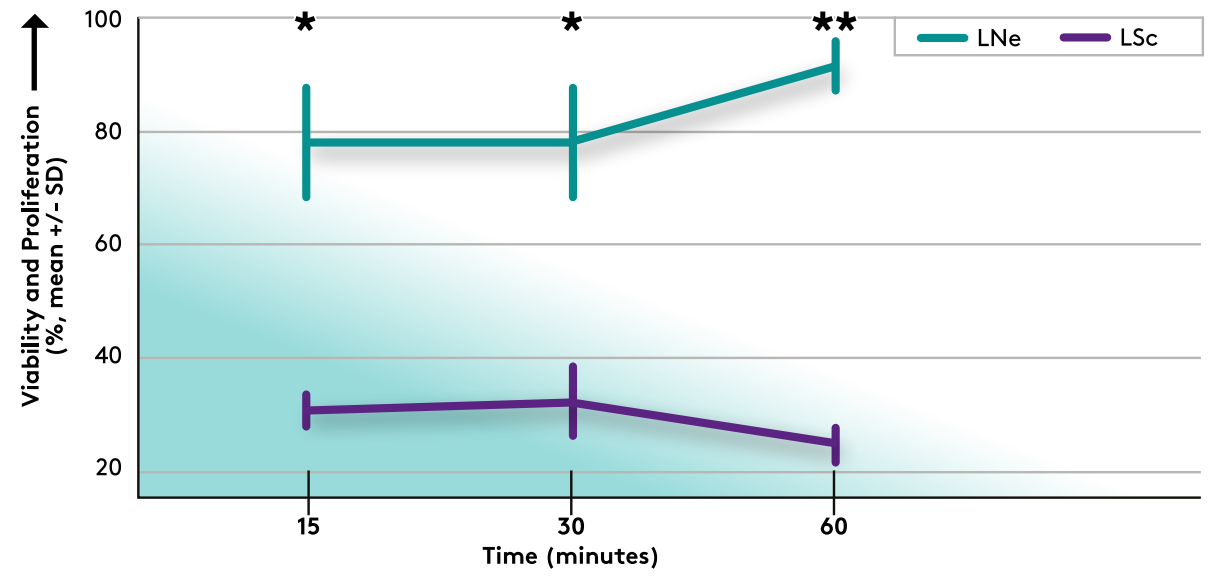
Viability and Proliferation (%, mean +/- SD)
100
80
60
40
20
15
30
60
Time (minutes)
*
*
**
LNe
LSc

Figure 3. Cytotoxicity evaluation by MTT assay. * p< 0.05; ** p< 0.01

CELL DEATH EVALUATION

Viable cells

The percentage of viability was significantly lower in cells exposed to LSc compared with those incubated with LNe at all study times.

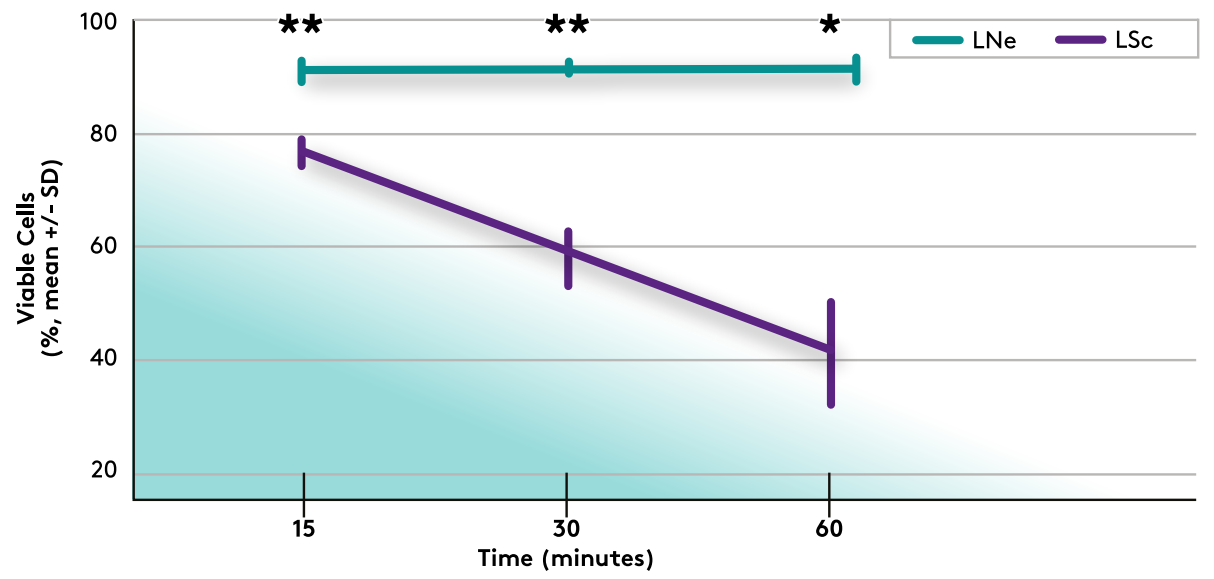
Viable Cells (%, mean +/- SD)
100
80
60
40
20
15
30
60
Time (minutes)
**
**
*
LNe
LSc

Figure 4. Percentage of viable cells (AV-/PI-) *P< 0.01; ** P< 0.05.

Early Apoptotic cells

No statistically significant differences were reached between groups in early apoptotic cells.

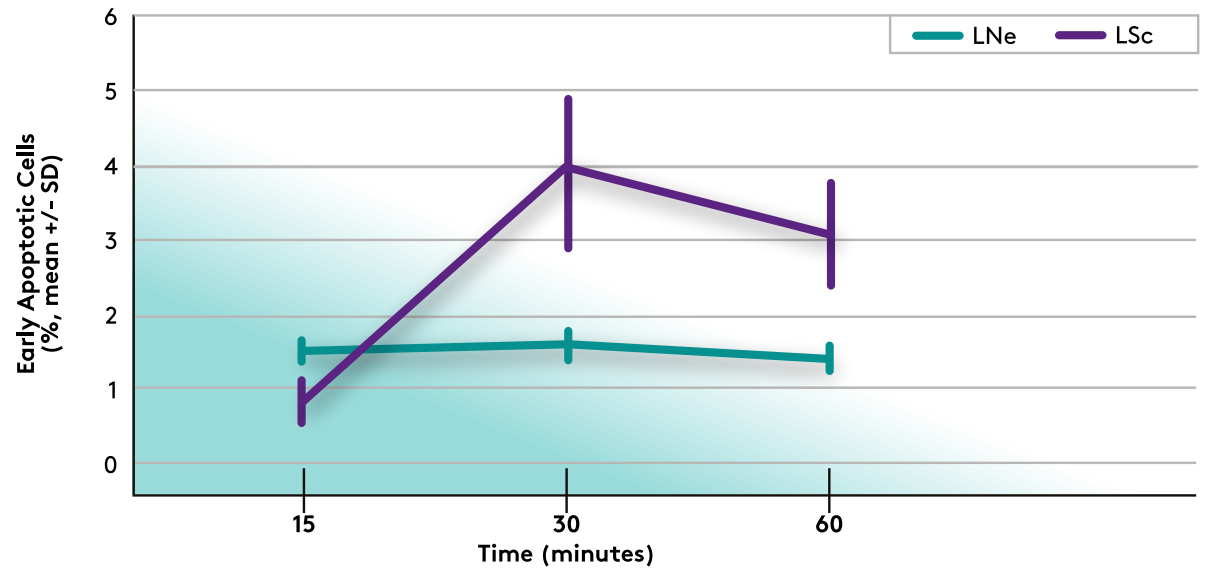
Early Apoptotic Cells (%, mean +/- SD)
6
5
4
3
2
1
0
15
30
60
Time (minutes)
LNe
LSc

Figure 5. Percentage of early apoptotic cells (AV+/PI-).

Late Apoptotic & Necrotic cells

A significant increase in late apoptosis and necrotic cells was observed at all study times for those incubated with LSc while LNe values remained around 5% at all study times.

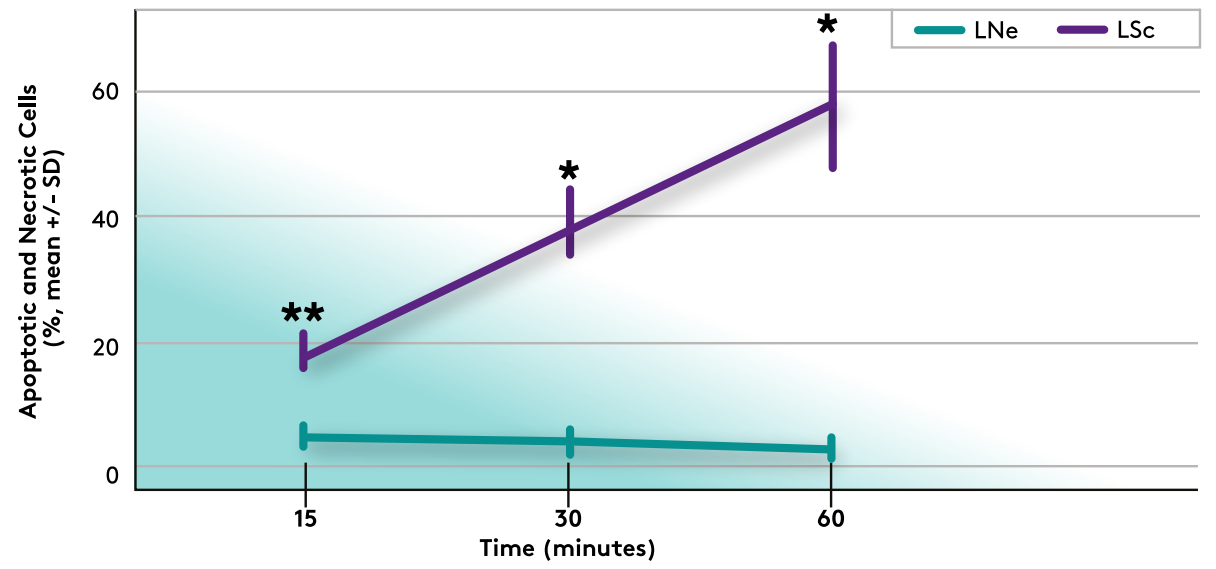
Apoptotic and Necrotic Cells (%, mean +/- SD)
60
40
20
0
15
30
60
Time (minutes)
**
*
*
LNe
LSc

Figure 6. Percentage of late apoptotic and necrotic cells (AV+/PI+). *P< 0.01; ** P< 0.05

CONCLUSION:

- **The new latanoprost nanoemulsion is significantly less cytotoxic on human conjunctival cells than latanoprost solution with BAK.**
- **This suggests that the new formulation could be gentler on the ocular surface than currently available BAK preserved latanoprost solutions, with levels of cytotoxicity equivalent to control.**

Acknowledgments:
Rodrigo M. Torres, MD PhD, for his scientific advice.

Disclosure:
This study was supported by POEN Laboratory.